AP5346 (ProLindac™), a pH-dependent polymer-vectorized DACH platinum, is active in borderline potentially platinum-sensitive ovarian cancer (OC) patients: results from an ongoing Phase I/II trial.

Florence Joly[1], Anne Madroszyk[2], Anne Floquet[3], Daniel Gedouin[4], Fabrice Bourdel[5], Mario Campone[6]

[1]Centre Francois Baclesse, Caen, France; [2]Institute Paoli Calmette CRLCC Marseille, France; [3]Institute Bergonié CRLCC Bordeaux, France; [4]Centre Eugène Marquis, Rennes, France; [5]AAIOncology, Le Kremlin Bicêtre, France; [6]Centre René Gauducheau CRLCC Nantes, France.

ABSTRACT

Background:
We have previously reported findings of an ongoing dose-intensity guided phase I/II trial with AP5346 (ProLindac™), a novel DACH-platinum, in patients with multi-treated ovarian cancer. In q2w and q3w schedules, treatment at 2 dose intensity levels (300 mg/m²/wk and 466 mg/m²/wk) was found to be well tolerated [1]. Here we provide updated results from the study, focusing on the most recent dose level (DL +2, DI=560 mg/m²/wk), in which as of May 2008, 5 patients (pts) have been treated (3 in Arm A, 2 in Arm B – q2w and q3w, respectively).

Material and Methods:
Pts having failed 2-4 prior chemotherapy (CT) lines were eligible provided they had ≥6 months of platinum-free progression-free interval (PPFI) (potentially platinum-sensitive), adequate organ function and evaluable (Rustin and/or RECIST) disease. Standard anti-emetic prophylaxis and hydration (2L NS with NaHCO3) were given before and after 1-hour ProLindac infusion.

Results:
From June 2006 until May 2008, 22 pts were enrolled in the first three dose levels (6 pts in DL0: 300 mg/m²/wk; 11 pts in DL+1: 466 mg/m²/wk; 5 pts in DL+2: 560 mg/m²/wk). Median age: 63 years (range: 45-70), median number of previous CT lines: 3 (2-7), and median PPFI is 17.4 months (6.9-42.6). Median Ca125 levels at baseline were 18.6x upper normal limit (UNL) for DL0, 6.3 UNL for DL+1pts, and 17.1 UNL for DL+2 pts. Median number of cycles (1 dosing per cycle) was only 2 (2-4) for DL0, since 5/6 pts had outright progression, 3 cycles (1-8) for DL+1, and 6 cycles (2-8) for DL+2 (3 pts ongoing). Safety: no renal toxicity or significant neutropenia or thrombopenia were reported; moderate nausea and vomiting were observed. Clinical delayed cisplatin-like neurotoxicity was seen in 3 pts (one with grade 2, two with grade 3), several weeks after 6, 3 and 3 cycles, respectively. All 4 evaluable pts to date treated in DL+2 (2 treated at q2w, 2 at q3w) have received ≥4 cycles (4/6/8/8 cycles), with 3/3 consistent Ca125 decreases (1 PR, 2 MR, Rustin criteria) and 1 SD (8 q2w cycles) in a pt with normal Ca125. Expanded accrual at the current DL (+2) is planned, with PK/PD and prospective neurotoxicity evaluation.

Conclusions:
The level of activity observed in the first pts of DL +2 compares favorably with published reports with oxaliplatin in the same population. Studies in combination with paclitaxel and gemcitabine in the same clinical setting, and in other indications, are planned for Q4 2008.

INTRODUCTION

Platinum-Based Therapy
Platinum-based drugs are among the most active anticancer agents and have been widely used in the treatment of a variety of human tumours. Although ovarian cancer is a chemosensitive malignancy, at least 20% of tumours are consistently resistant to first-line platinum-based chemotherapy. Chemoresistance in advanced ovarian cancer (AOC) is at least partially related to DNA mismatch-repair (MMR) deficiency, which has been reported in 5-25% of primary AOC and in over 60% of platinum pre-treated tumours.

Oxaliplatin is a DACH platinum analogue with a mechanism of action similar to classic platinum compounds. Preclinical data however, show that DACH-platinum-DNA adducts are bulkier and more hydrophobic than cis-diamine-platinum-DNA adducts and perhaps more effective in DNA synthesis inhibition, causing greater cytotoxicity. In particular, the MMR enzymatic complex does not recognize DACH-platinum-DNA adducts, which can partially explain the lack of cross-resistance to oxaliplatin in cisplatin-resistant cell lines.

AP5346
AP5346 is a novel hydrophilic biocompatible copolymer to which DACH-platinum is attached. This pH-dependent polymer prodrug is designed to improve the efficacy and safety profile of the DACH-platinum by taking advantage of the hyper-permeable vasculature of tumours, thus increasing the concentration of the drug at the hypoxic/acidic tumor site. The differential biotransformation profile and sustained release of platinum from the polymer may improve safety compared to oxaliplatin and cisplatin.

Clinical Data
In an initial phase I trial AP5346 was administered as a 1-hour infusion on days 1, 8 and 15 of a 28-day cycle [2]. AP5346 was shown to be an active compound, with clinically meaningful responses seen at a recommended dose (RD) of 640 mg/m² in heavily pretreated ovarian cancer patients. Results showed AP5346 to be well tolerated. Toxicities (nausea and vomiting) occurred frequently in pre-treated patients in a dose dependant manner, but were non-life-threatening and usually reversible. At doses exceeding the RD, some severe hematological toxicity (neutropenia/thrombocytopenia), and renal toxicity related morbidities were seen.

This Study
We decided to further explore dosing and scheduling optimization in a phase II setting using a more convenient d1 q3w schedule with a dose intensity guided escalation, using potentially sensitive ovarian cancer patients as the target population. In an amendment to the original protocol design, following treatment of patients at the first DL without evidence of toxicity or activity, the dose finding scheme was expanded to include higher dose intensities than originally planned and to investigate an alternative dosing schedule of treatment q2w.

AIMS OF THE STUDY

Primary Objective
- To evaluate antitumor activity (RECIST and GCIC-Rustin modified criteria for CA125 response [3, 4]) of single agent AP5346 as 3rd or 4th line chemotherapy in AOC patients, previously pre-treated with organoplatins (except oxaliplatin) + taxanes + anthracyclines with a platinum-free, progression-free interval (PPFI) of ≥ 6 months.

Secondary Objectives
- To optimize the dose of intravenous AP5346 administered every 3 weeks.
- To evaluate safety.
- To evaluate antitumor activity in terms of response duration, progression free survival and overall survival.
- To investigate the pharmacokinetics AP5346 in this patient population.

STUDY DESIGN

This is an open-label, single arm, multicenter, phase I/II trial with AP5346 administered as a 2-hour intravenous infusion to AOC patients on day 1 of a 2-week or 3-week cycle.

Dose escalation scheme
Treatment started at 900 mg/m², on a d1 q3w schedule. This schedule has a dose intensity of 300 mg/m²/week, equivalent to 66% of the dose intensity (DI) of the RD from the previous phase I study[2] (RD=480 mg/m²/week, based on 640 mg/m²/week, d1,8,15 q4w).
The dose escalation scheme is based on 2 stages, and is presented in the following diagram. Dose escalations were based on DI; in the second treatment regimen (regimen B, q2w), doses were selected to provide the same DI as the equivalent dose level in regimen A (q3w).
Doses represent milligrams of platinum administered.

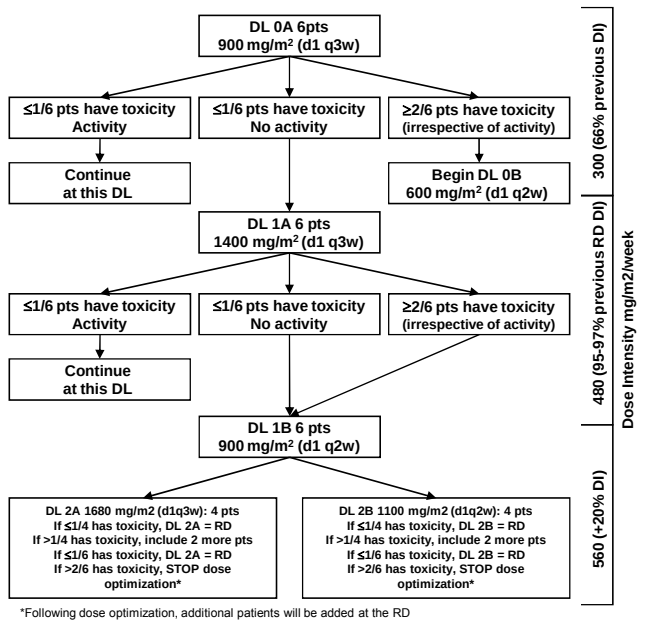
DL 0A 6pts
900 mg/m² (d1 q3w)
≤1/6 pts have toxicity Activity
≤1/6 pts have toxicity No activity
≥2/6 pts have toxicity (irrespective of activity)
Continue at this DL
Begin DL 0B 600 mg/m² (d1 q2w)
DL 1A 6 pts
1400 mg/m² (d1 q3w)
≤1/6 pts have toxicity Activity
≤1/6 pts have toxicity No activity
≥2/6 pts have toxicity (irrespective of activity)
Continue at this DL
DL 1B 6 pts
900 mg/m² (d1 q2w)
DL 2A 1680 mg/m2 (d1q3w): 4 pts
If ≤1/4 has toxicity, DL 2A = RD
If >1/4 has toxicity, include 2 more pts
If ≤1/6 has toxicity, DL 2A = RD
If >2/6 has toxicity, STOP dose optimization*
DL 2B 1100 mg/m2 (d1q2w): 4 pts
If ≤1/4 has toxicity, DL 2B = RD
If >1/4 has toxicity, include 2 more pts
If ≤1/6 has toxicity, DL 2B = RD
If >2/6 has toxicity, STOP dose optimization*
300 (66% previous DI)
480 (95-97% previous RD DI)
560 (+20% DI)
Dose Intensity mg/m2/week

*Following dose optimization, additional patients will be added at the RD

Eligibility Criteria
1. Patients with recurrent platinum-sensitive OC previously treated with platinum-based chemotherapy (except oxaliplatin) + taxane as 1st line and anthracyclines as 2nd or 3rd line. The platinum-based treatment interval (PFI) must be ≥6 months provided that there was no progression during or within 3 months of such discontinuation.
2. Measurable/evaluable disease (lesions or CA 125 levels)
3. Age ≥18 years; life expectancy ≥ 3 months, signed informed consent; ECOG Performance Status (PS): 0-1
4. Adequate hematological and biological function

Patients were to be excluded if:
1. ≥6 prior chemotherapy lines* (including both on- and off-label active agents in the ovarian cancer indication)
2. ≥3 previous platinum-based regimens (cisplatin and/or carboplatin)
3. Prior treatment with oxaliplatin
4. Antecedent of NCI-CTCAE > grade 3 hypersensitivity reaction to organoplatins

***Given the plethora of single agent regimens used in this indication, a recent protocol amendment has increased the number of permissible prior chemotherapy lines to from three to five.**

RESULTS

Patient Characteristics
Between June 2006 and September 2008, 26 patients have been included and treated.

Table 1. Patient Characteristics

Dose Level (mg/m^2)	DL 0 900 q3w	DL+1A 1400 q3w	DL+1B 900 q2w	DL+2A 1680 q3w	DL+2B 1100 q2w	Total
N Patients	6	6	5	5	4	26
Age (years)						
Median	56	63	61	67	63	63
Range	45-69	51-68	50-63	50-70	60-73	45-73
Performance Status (ECOG)						
0	1	3	5	2	2	13
1	5	3	-	3	2	13
N organs involved at baseline						
Median	2	3	2	2	3	2
Range	1-4	1-4	1-2	1-3	2-4	1-4
CA125 at baseline (xULN)						
Median	548	156	386	127	1166	277
Range	16-1854	69-1910	105-1684	14-1269	43-2272	14-2272
N prior chemotherapy regimens						
Median	3	3	3	4	3.5	3
Range	2-4	2-4	2-6	2-4	2-7	2-7
N prior platinum regimens (cis-/carboplatin)						
Median	2	2	2	2	2	2
Range	1-3	1-4	1-3	1-3	2-3	1-4
PPFI under last Pt therapy (months)						
Median	8.3	13.6	4.2	6.8	6.8	7.3
Range	7.0-12.6	0.3-16.6	1.6-14.8	5.1-9.9	0.6-9.0	0.3-16.6

Safety
No patients experienced dose limiting toxicity (DLT; haematological toxicity ≥ grade 3 or clinically meaningful non-haematological toxicity) during this study. The most frequently reported related non-haematological toxicities were nausea (20 patients, including one grade 3) vomiting (10 patients, including one grade 3) and asthenia (10 patients, including one grade 3). Anaemia was reported in 2 patients – 1 grade 2, 1 grade 3 (Table 3).

Mild renal toxicity was reported in one patient although this patient reported abnormal renal function (creatinine clearance <60 mL.min-1) at baseline. Other patients showed little change in renal function over the course of treatment, as measured by creatinine clearance.

Three patients experienced SAE as follows:

One patient from the DL+1A treatment group experienced grade 4 hypersensitivity in the fifth cycle of treatment. This patient also experienced paraesthesia (grade 3) during follow-up.

A second patient in the same treatment group experienced grade 3 paraesthesia, balance disorder, vertigo and tinnitus (grade 2) during follow-up.

The third patient from the DL+2A treatment group experienced grade 3 vomiting during their first cycle of treatment, which was later ascribed to the development of an abdominal mass.

Table 2. Treatment exposure and reasons for discontinuation

Dose Level (mg/m^2)	DL 0 900 q3w	DL+1A 1400 q3w	DL+1B 900 q2w	DL+2A 1680 q3w	DL+2B 1100 q2w	Total
N Patients	6	6	5	5	4	26
N Cycles received						
Median	2	4	4	4	3.5	3.5
Range	2-4	2-5	2-8	2-9	1-8	1-9
Cumulative dose (mg/m^2)						
Median	1800	5600	3600	6720	3850	3585
Range	1718-3570	2839-7018	1800-7240	3360-15108	1100-8800	1100-15108
RDI						
Median	0.98	0.99	1.00	1.00	1.00	1.00
Range	0.86-1.0	0.82-1.0	0.79-1.0	0.38-1.0	1.0-1.0	0.38-1.0
Patients still on treatment	-	-	-	-	1	1
Reason for treatment discontinuation						
Related AE	-	1	-	2	-	3
PD	6	3	5	2	2	18
Investigator decision	-	2	-	1	1	4

Table 3. Frequency of Grade 3-4 Related Adverse Events by Dose Level (MEDdra)

Dose Level (mg/m^2)	DL 0 900 q3w	DL+1A 1400 q3w	DL+1B 900 q2w	DL+2A 1680 q3w	DL+2B 1100 q2w	Total
Blood and lymphatic system disorders						
Anemia	-	1	-	-	-	1
Neutropenia	-	1	-	1	-	2
Gastrointestinal disorders						
Nausea	-	-	-	-	1	1
Vomiting	-	-	-	-	1	1
Immune system disorders						
Hypersensitivity	-	1	-	-	-	1
Nervous system disorders						
Dysaesthesia	-	-	-	1	-	1
Paraesthesia	-	2	-	1	-	3

Ten patients experienced a nervous system disorder of any grade (Table 4) and four instances of grade 3 platinum-like cumulative neurotoxicity were described (Table 5): Two patients from the DL+1A regimen experienced grade 3 paraesthesia in their hands and feet during follow-up after 4 and 5 cycles of treatment. One of them also experienced a balance disorder (grade 2); this patient entered the study with diabetes and these AE were believed to be diabetes related neuropathies. Two other patients from the DL+2A regimen experienced either lower limb paraesthesia during the sixth treatment cycle or hand and foot dysaesthesia after nine cycles of treatment. None of these patients had received prior treatment with cisplatinum regimens. Other neurotoxic events were mild to moderate in intensity.

Table 4. Frequency of Nervous System Disorders, Worst Grade by Patient

Dose Level (mg/m^2)	DL 0 900 q3w				DL+1A 1400 q3w				DL+1B 900 q2w				DL+2A 1680 q3w				DL+2B 1100 q2w				Total
Grade	1	2	3	4	1	2	3	4	1	2	3	4	1	2	3	4	1	2	3	4	
Areflexia	1	-	-	-	-	-	-	-	-	-	-	-	-	-	-	-	-	-	-	-	1
Balance disorder	-	-	-	-	-	1	-	-	-	-	-	-	-	-	-	-	-	-	-	-	1
Dysaesthesia	-	-	-	-	-	-	-	-	-	-	-	-	-	-	1	-	-	-	-	-	1
Paraesthesia	1	-	-	-	1	-	2	-	-	-	-	-	-	-	1	-	-	-	-	-	5
Peripheral sensory neuropathy	-	-	-	-	-	-	-	-	1	-	-	-	-	-	-	-	-	-	-	-	1

Table 5. Frequency of Neurotoxicity by Total Dose Received

Dose Intensity:	0-2000 mg/m^2	2000-5000 mg/m^2	>5000 mg/m^2
N Patients	8	9	9
Neurotoxicity-grade			
1	2*	1	-
2	-	-	-
3	-	-	4

*** One patient had concomitant diabetic neuropathy**

Efficacy
The best overall response recorded by any patient was SD (16 patients, 62%) (Table 6). For one patient in the DL+2B treatment group it is too early to measure a response. One patient experienced a reduction in CA125 levels which qualified a response to treatment according to GCIC Rustin modified criteria, although several patients, particularly at DL+2 experienced clinically meaningful CA125 stabilizations (Figure 1 and Figure 2).

Table 6. Overall Tumor Response (RECIST / GCIC-Rustin modified criteria)

	DL 0 900 q3w	DL+1A 1400 q3w	DL+1B 900 q2w	DL+2A 1680 q3w	DL+2B 1100 q2w	Total
N evaluable pts	5	5	5	4	2	21
Best-response						
SD	1	4	3	2	2	12
PD	4	1	2	2	-	9
Non evaluable	1	1	-	1	2	5
Pts evaluable for CA125	0	2	5	5	3	15
CA125 response	-	-	-	1	-	1

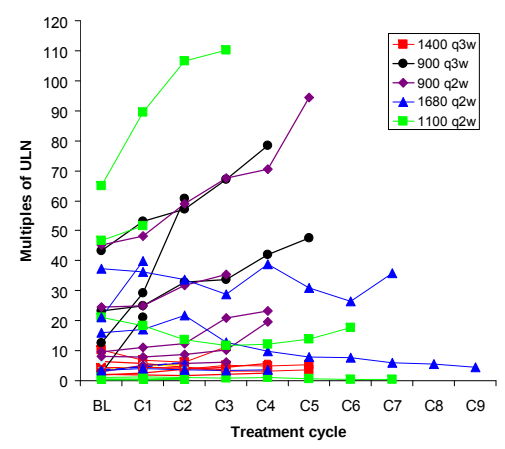
1400 q3w
900 q3w
900 q2w
1680 q2w
1100 q2w
Multiples of ULN
0
10
20
30
40
50
60
70
80
90
100
110
120
BL
C1
C2
C3
C4
C5
C6
C7
C8
C9
Treatment cycle

Figure 1. Serum levels of CA125 over time by treatment group
Each line of t

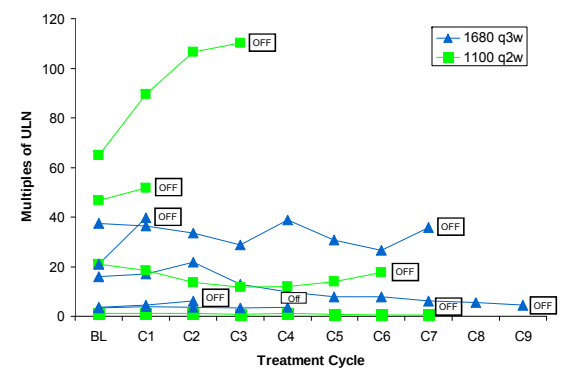
1680 q3w
1100 q2w
Multiples of ULN
0
20
40
60
80
100
120
OFF
Off
BL
C1
C2
C3
C4
C5
C6
C7
C8
C9
Treatment Cycle

Figure 2 . Serum levels of CA125 over time by treatment group (DL+2)
Each line of the same color represents a different patient from the same treatment cohort.

CONCLUSIONS

Despite the heavily pre-treated nature and clinical resistance to platinum drugs of the study population, sustained levels of activity were seen in several patients at higher dose levels. Furthermore, the level of activity observed in the patients treated at DL +2 benchmarks favorably with previous published reports of single agent oxaliplatin in a similar but less pre-treated population.
According to the standard phase I study design, the MTD / RD based on acute or sub acute phenomena, were not reached. There were nevertheless consistent signs of delayed cumulative neurotoxicity after multiple cycles and a total dose of >5 g. As a result and because of several meaningful stabilizations and Rustin criteria responses, it was decided that DL +2 for either regimen (1680 mg/m^2 q3w or 1100 mg/m^2 q2w) is to be the RD for further studies.
The imminent availability of large scale GMP batches of AP5346 (rather that the midsized batches used currently) and new insights into the protein binding characteristics of AP5346 and platinum release rate (see Poster 104) will necessitate the re-evaluation of DL 1 and 2 with our new pharmacokinetic assessment methodology in a similar patient population. Studies in combination with paclitaxel, gemcitabine and flouropyrimidines in the same clinical setting, and in other indications, are also planned for Q4 2008.

REFERENCES

[1] Campone M., Floquet MA., *et. al.* (2007) AP5346, a pH-dependent polymer vectorized DACH, is safe at pharmacodynamically active doses: results from an ongoing Phase II trial in potentially platinum-sensitive ovarian cancer (OC) patients. EORTC-NCI-AACR, 2007

[2] Campone M, Rademaker-Lakhai J, Bennouna J, *et al* (2007) Phase I and pharmacokinetic trial of AP5346, a DACH-platinum-polymer conjugate, administered weekly for three out of every 4 weeks to advanced solid tumor patients. *Cancer Chemother Pharmacol* 60:523-533.

[3] Therasse P, Arbuck SG, Eisenhauer EA, *et. al.* (2000) New guidelines to evaluate the response to treatment in solid tumors. European Organization for Research and Treatment of Cancer, National Cancer Institute of the United States, National Cancer Institute of Canada. J Natl Cancer Inst 92:205-216.

[4] Rustin G, Quinn M, Thigpen T, *et. al.* (2004) Re: New Guidelines to Evaluate the Response to Treatment in Solid Tumors (Ovarian Cancer). J Natl Cancer Inst 96:487-488.